Exploratory Study of Continuous Glucose Monitoring in the Epidural Space in Swine

Paul V Goode, PhD; Michael Talcott, DVM; JP Thrower, PhD; Timothy L Routh, M Eng.; Stephen T Tapsak; Samantha Wakil, PhD; Mark A Tapsak, PhD | Corresponding author: Mark A Tapsak; mtapsak@glucotrack.com

Background:

Glucose can be measured in various compartments of the body. This first-of-its-kind exploratory study evaluated epidural sensing as a viable method for continuous glucose monitoring.

Objective:

The primary objectives were to evaluate intra- and inter-device performance, assess the suitability of sensing in the epidural space, and examine chronic biocompatibility and histology through explant and necropsy.

Methods:

A preclinical study was conducted using a chronic porcine model to evaluate a novel long-term implant designed for the epidural space. The test device was comprised of an off-the-shelf spinal cord stimulation (SCS) lead that had been minimally modified by placing glucose-sensing membranes atop 4 of the 8 existing stimulation electrodes. The resulting sensing lead and subcutaneous housing units were implanted in 4 swine, for up to 56 days. Glucose levels were continuously monitored using the test device. Periodic glucose excursions were induced through oral glucose tolerance tests (~136 g) and intravenous glucose tolerance tests (~32 g). Glucose measurements from the epidural space were compared with those obtained from blood and interstitial fluid using commercially available devices.

CGM=continuous glucose monitor; IVGTT=intravenous glucose tolerance test; MARD=mean absolute relative difference; OGTT=oral glucose tolerance test.

Results: Glucotrack data is unfiltered

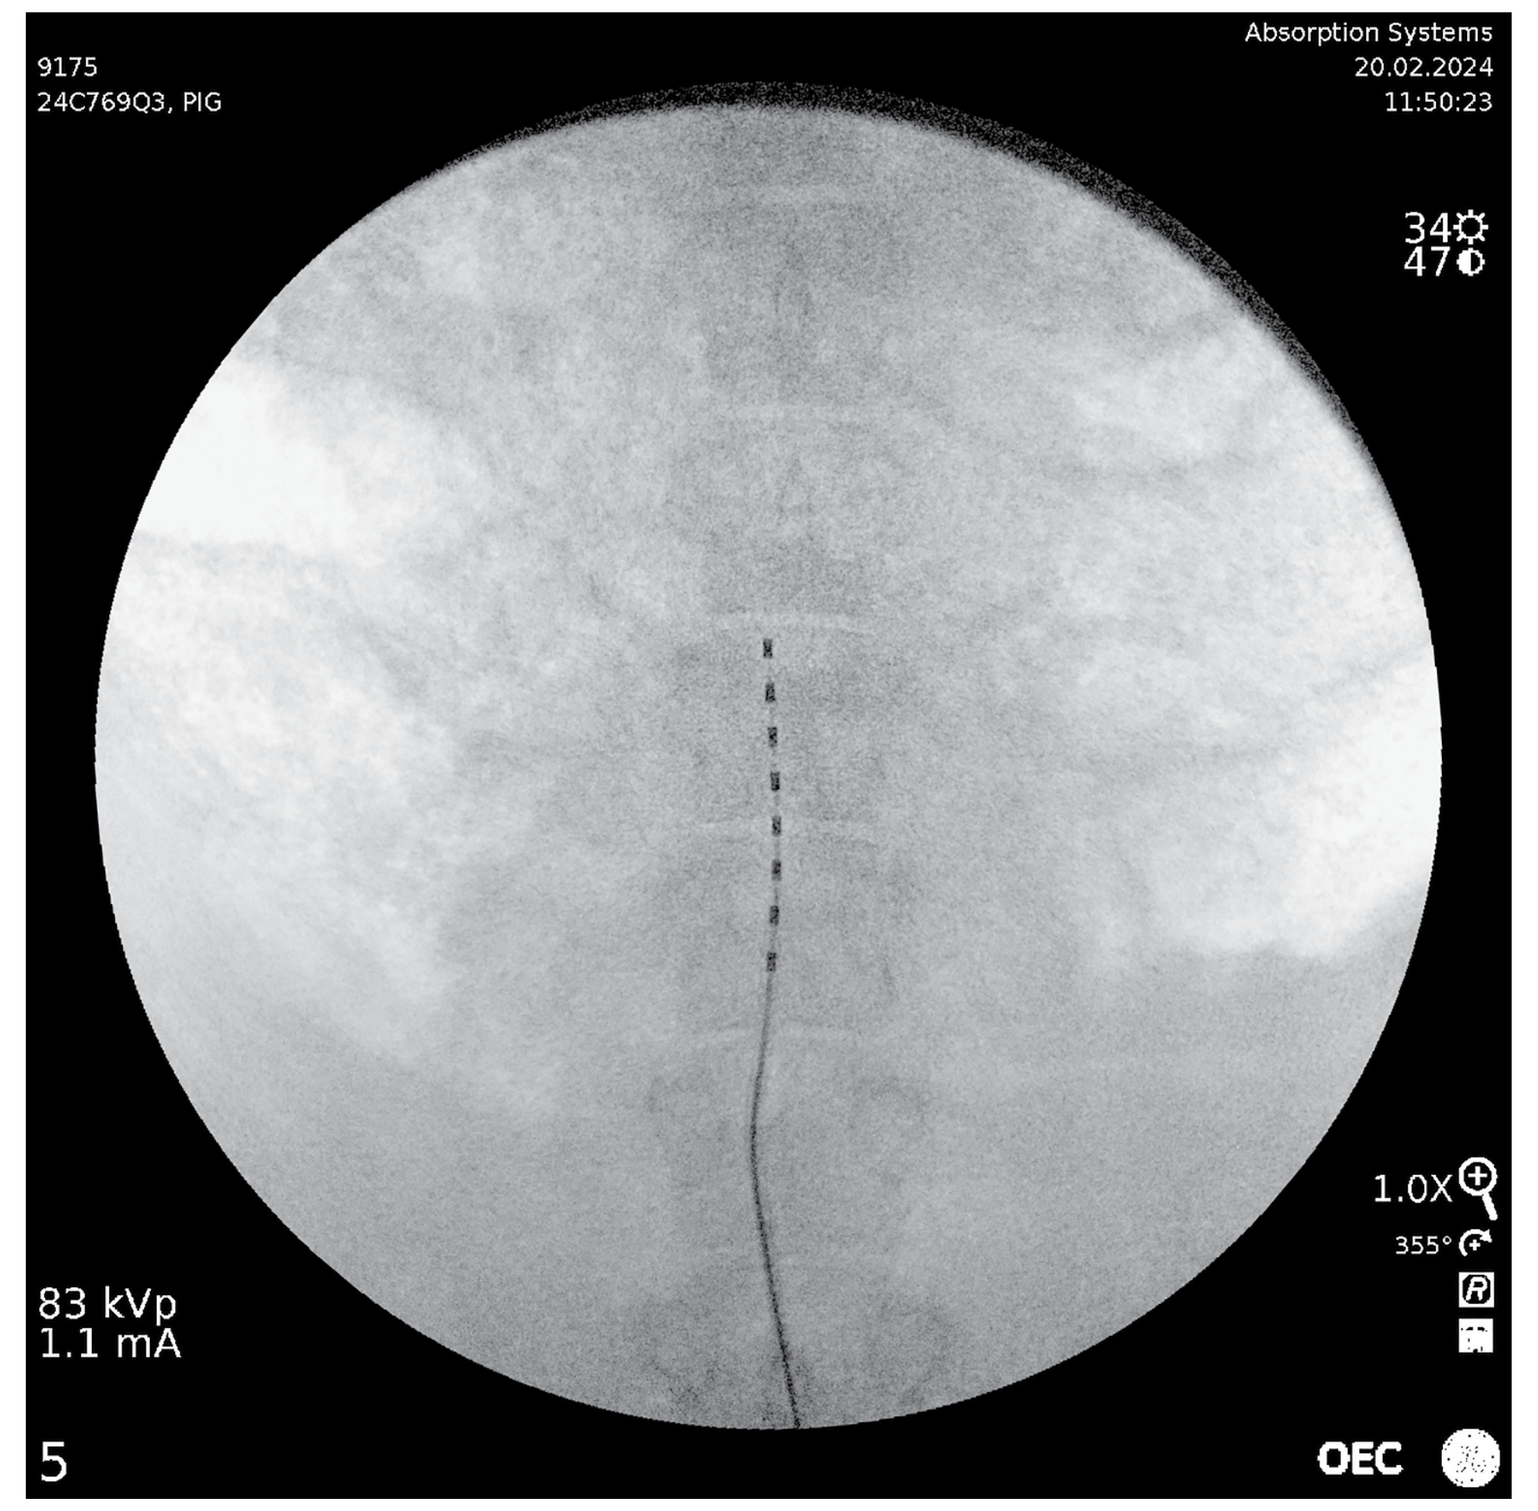
9175
24C769Q3, PIG
Absorption Systems
20.02.2024
11:50:23
83 kVp
1.1 mA
1.0X
OEC

Figure A: Example lead placement within the epidural space.

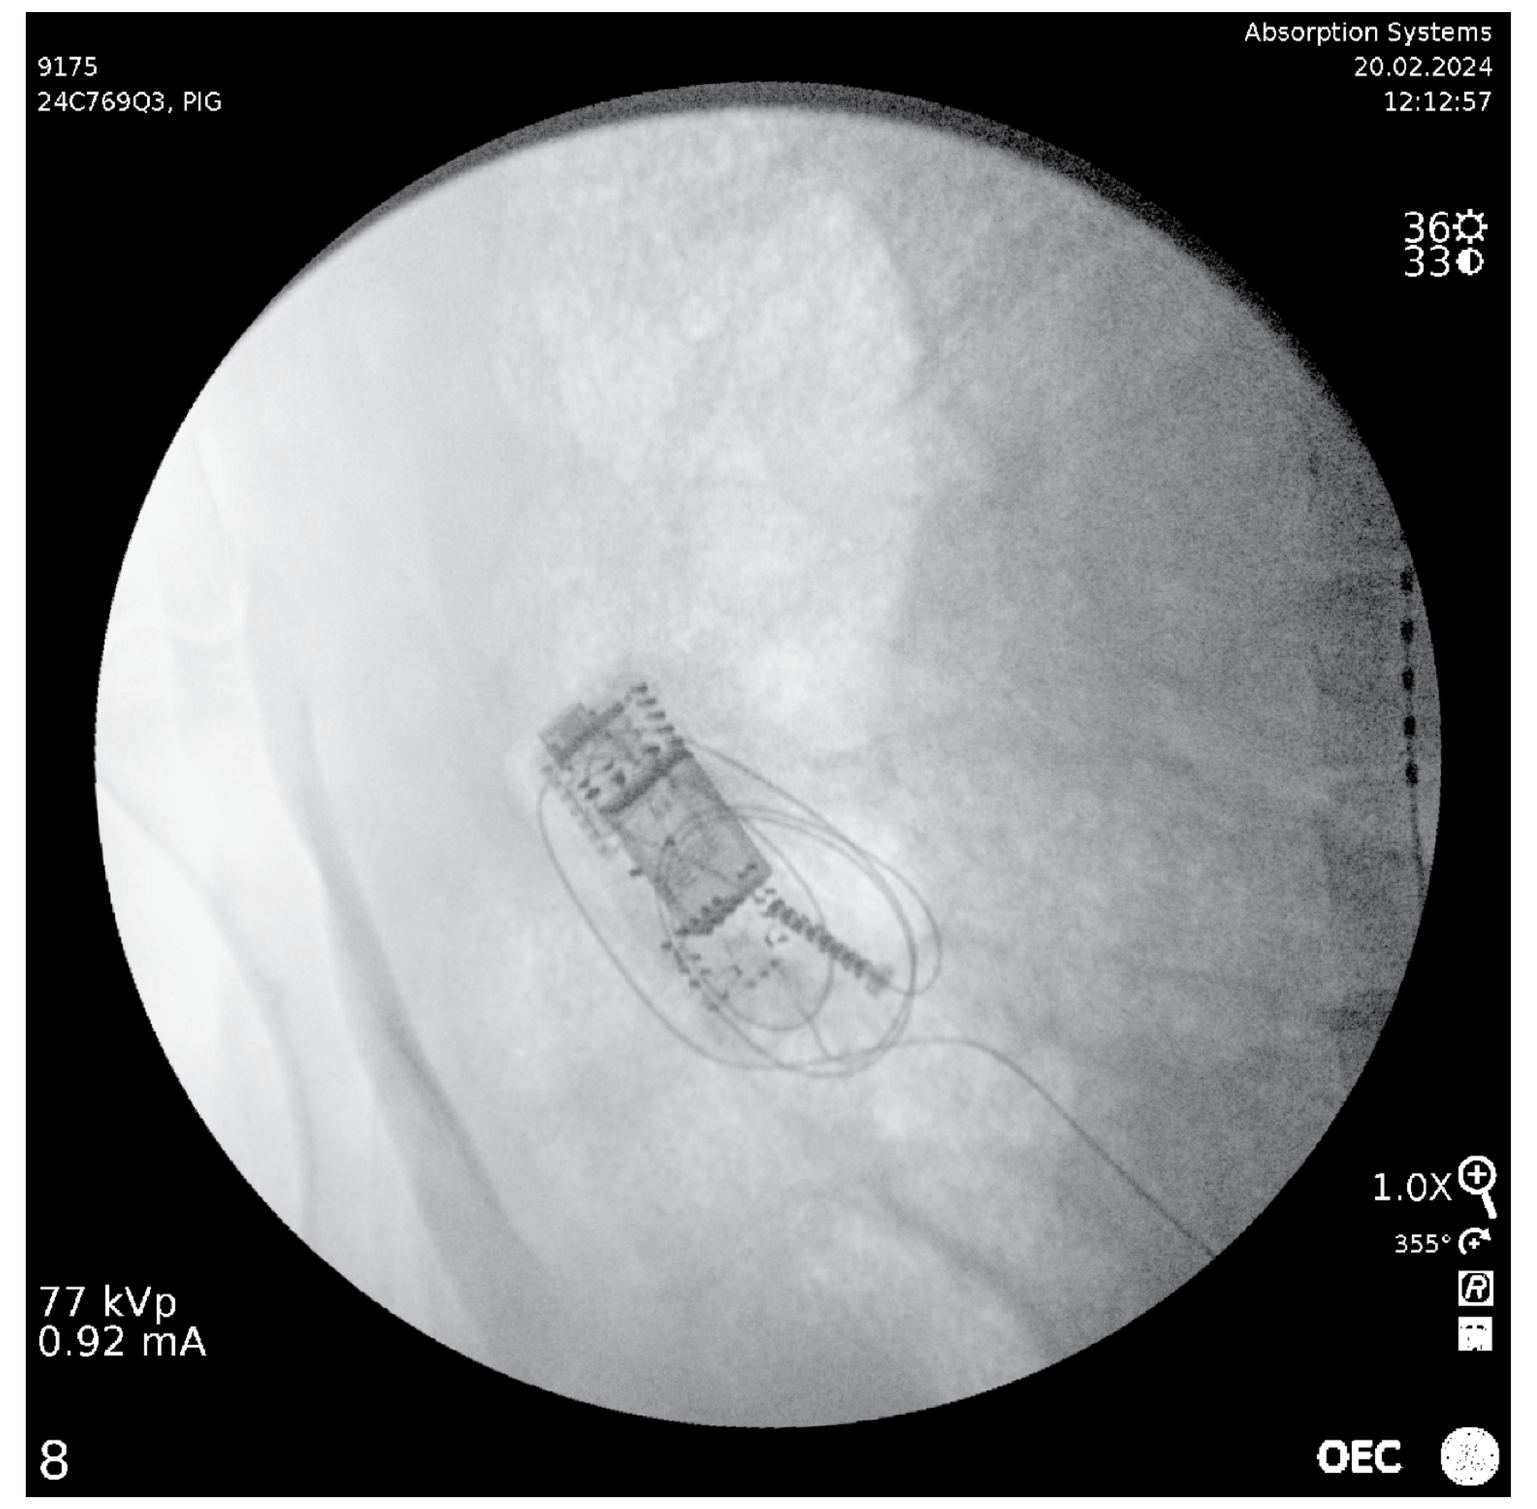
9175
24C769Q3, PIG
Absorption Systems
20.02.2024
12:12:57
77 kVp
0.92 mA
1.0X
OEC

Figure B: Example of prototype animal device implanted in subcutaneous pocket (includes extra lead length).

Animal 1—24 Hours Post-Implant, OGTT; MARD = 5.0%

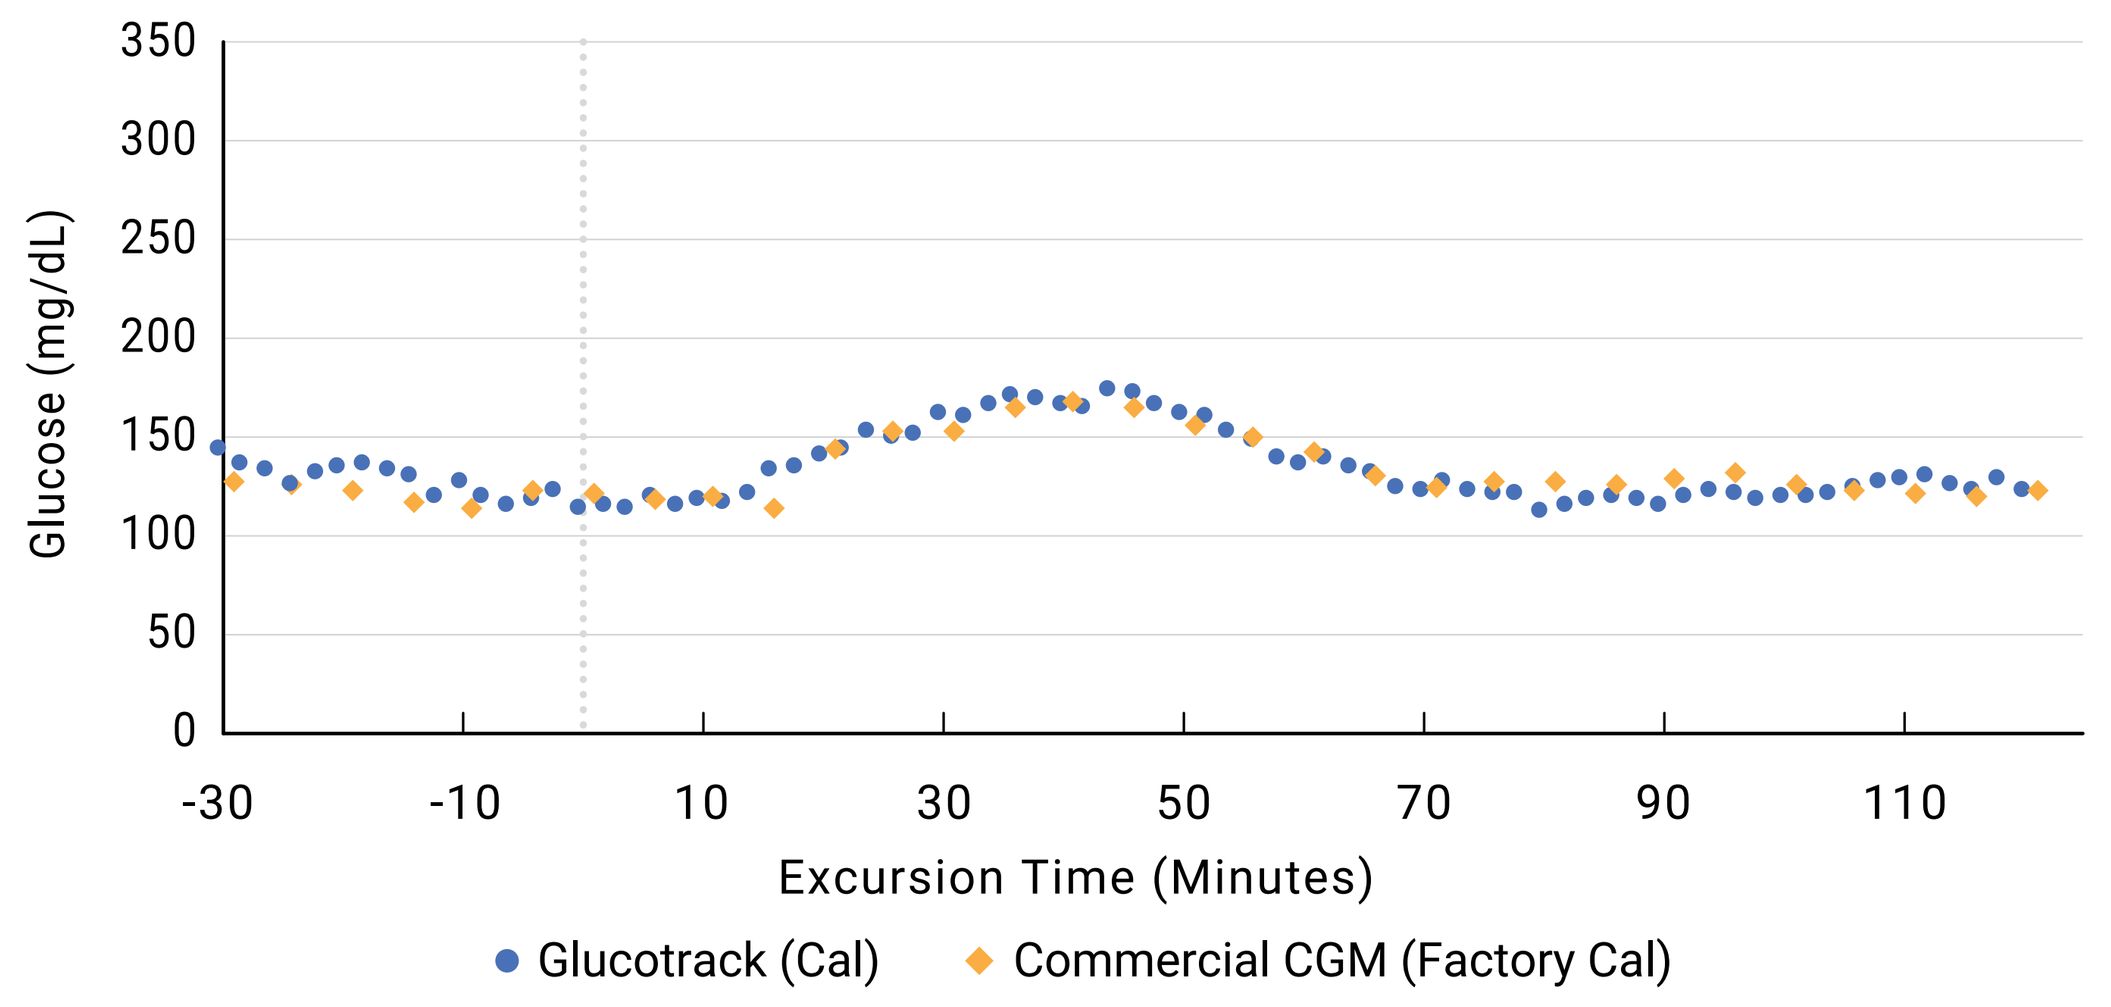
Glucose (mg/dL)
Excursion Time (Minutes)
Glucotrack (Cal)
Commercial CGM (Factory Cal)

Animal 2—Week 4, IVGTT; MARD = 12.9%

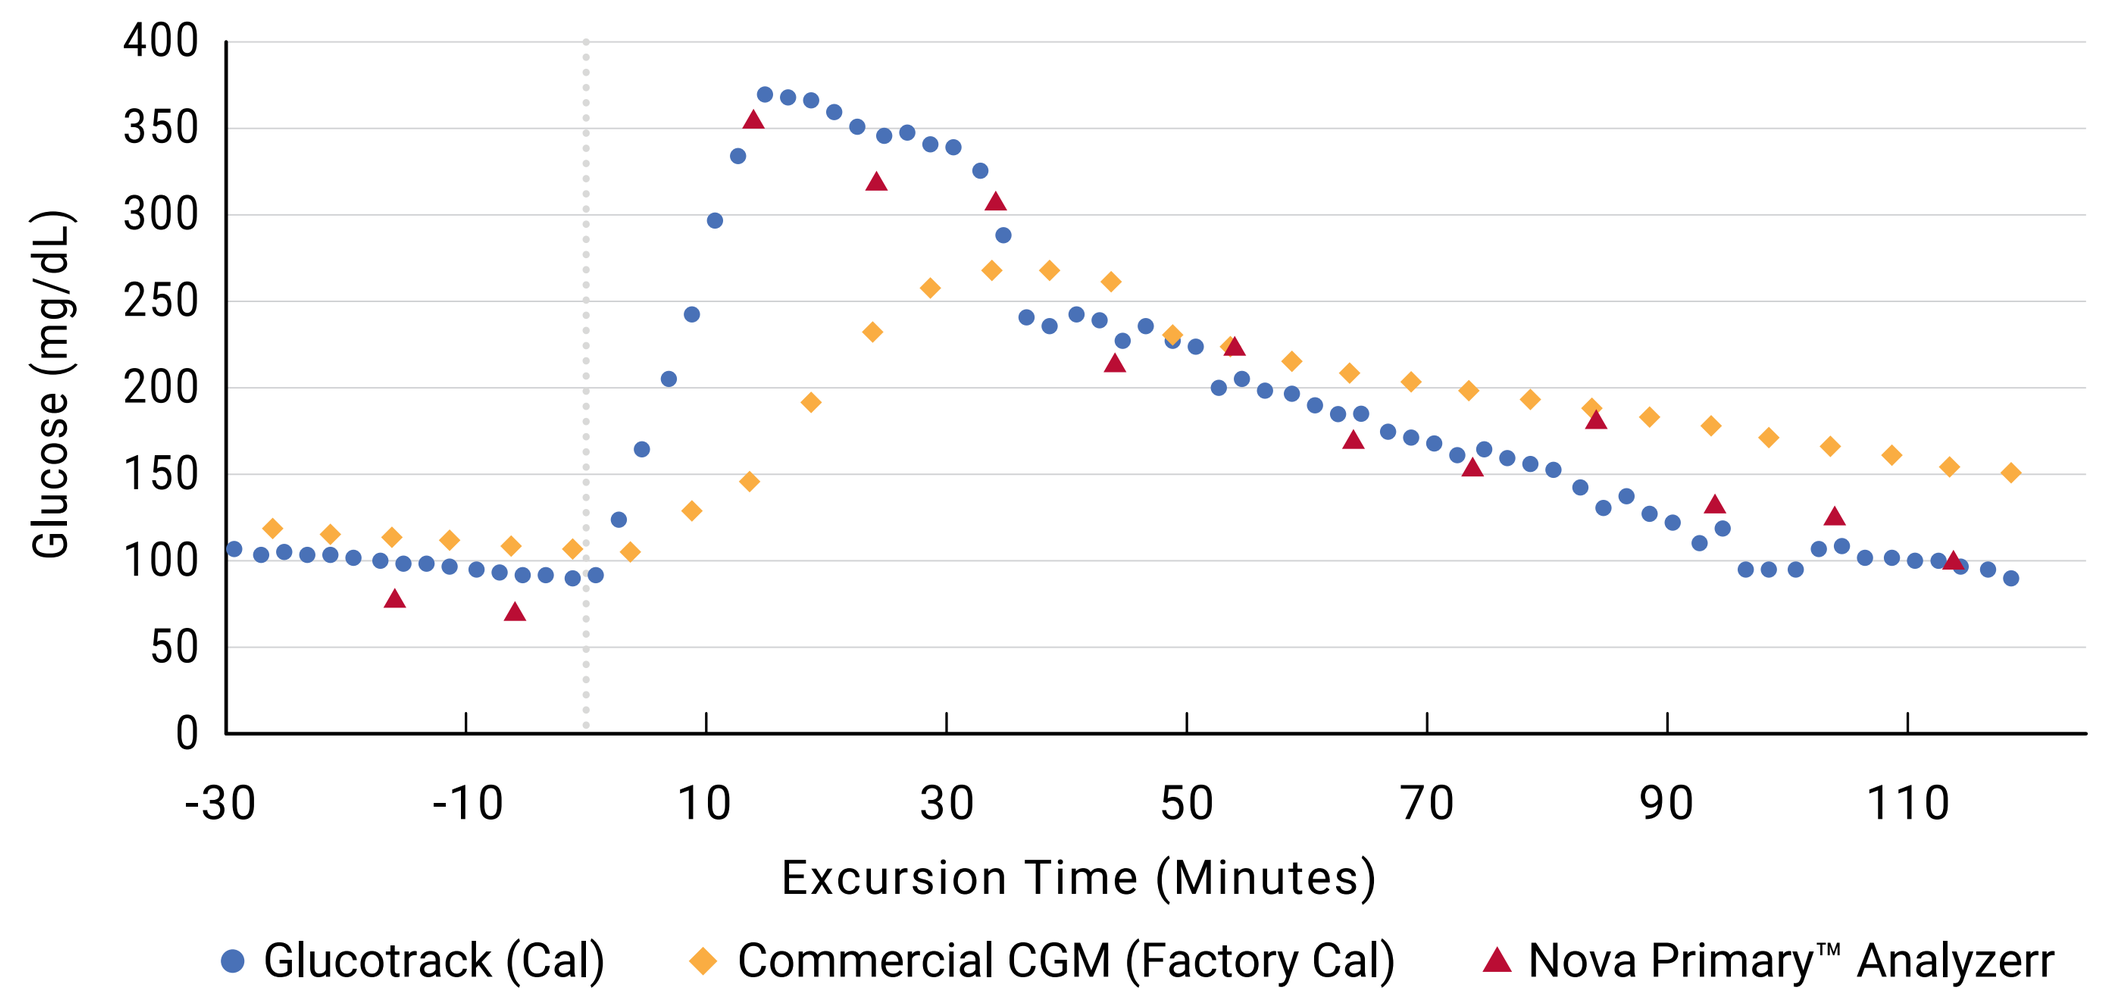
Glucose (mg/dL)
Excursion Time (Minutes)
Glucotrack (Cal)
Commercial CGM (Factory Cal)
Nova Primary™ Analyzerr

Animal 1—Week 3 OGTT; MARD = 3.0%

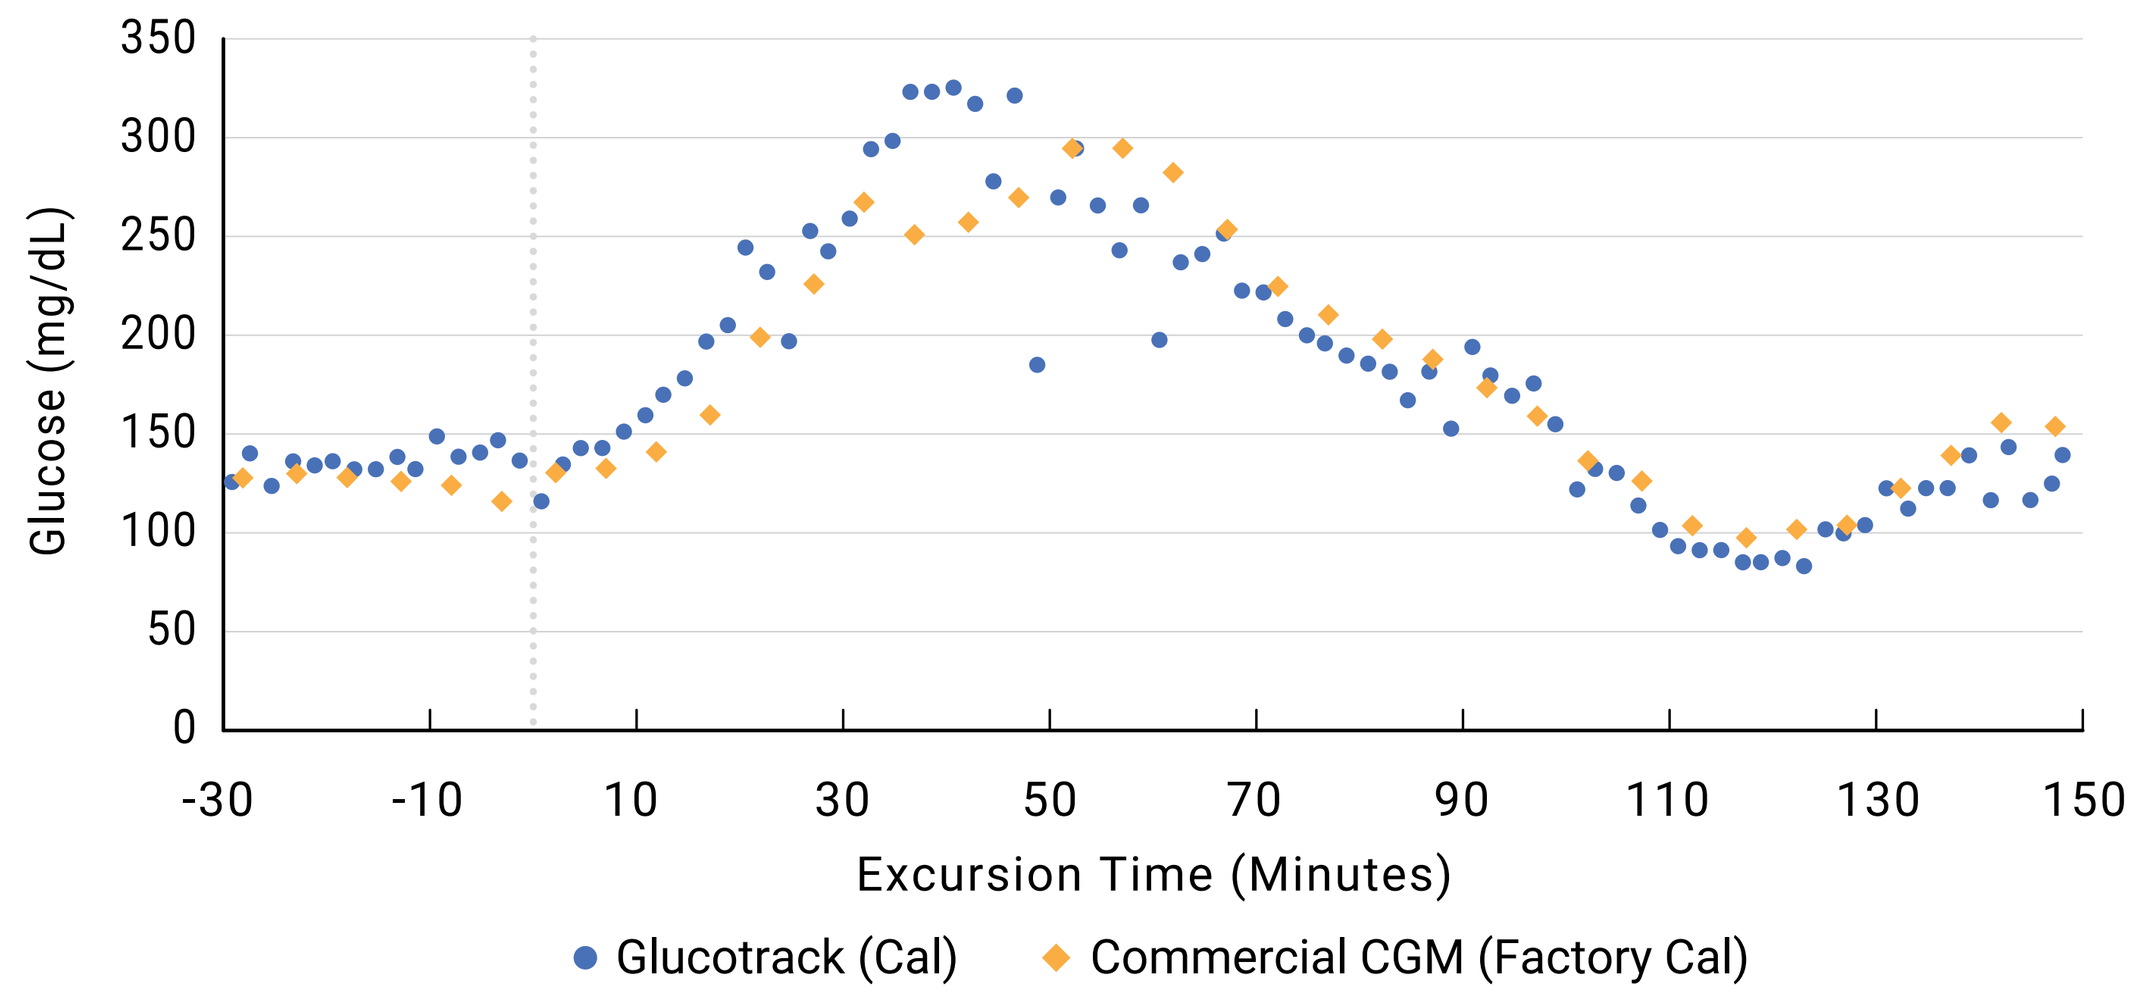
Glucose (mg/dL)
Excursion Time (Minutes)
Glucotrack (Cal)
Commercial CGM (Factory Cal)

Animal 1—Week 7, OGTT; MARD = 13.1%

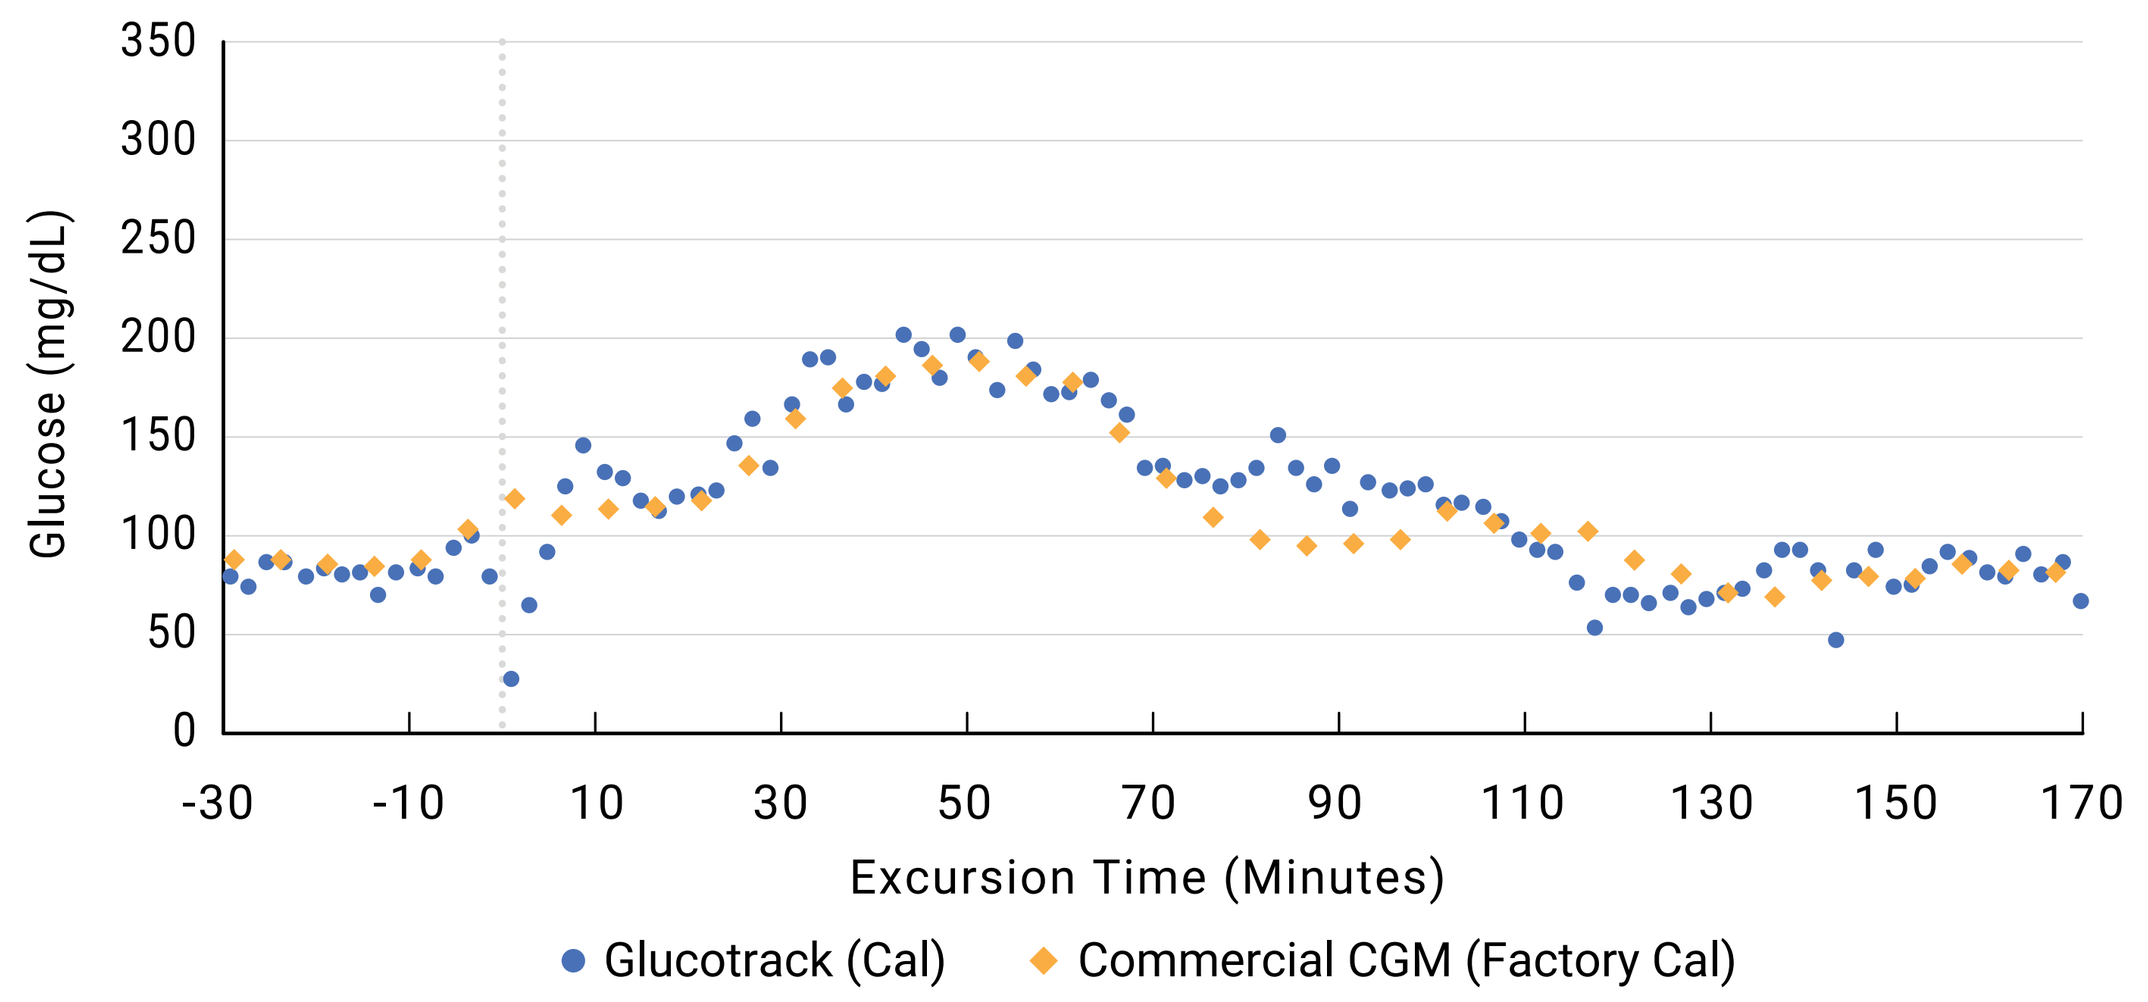
Glucose (mg/dL)
Excursion Time (Minutes)
Glucotrack (Cal)
Commercial CGM (Factory Cal)

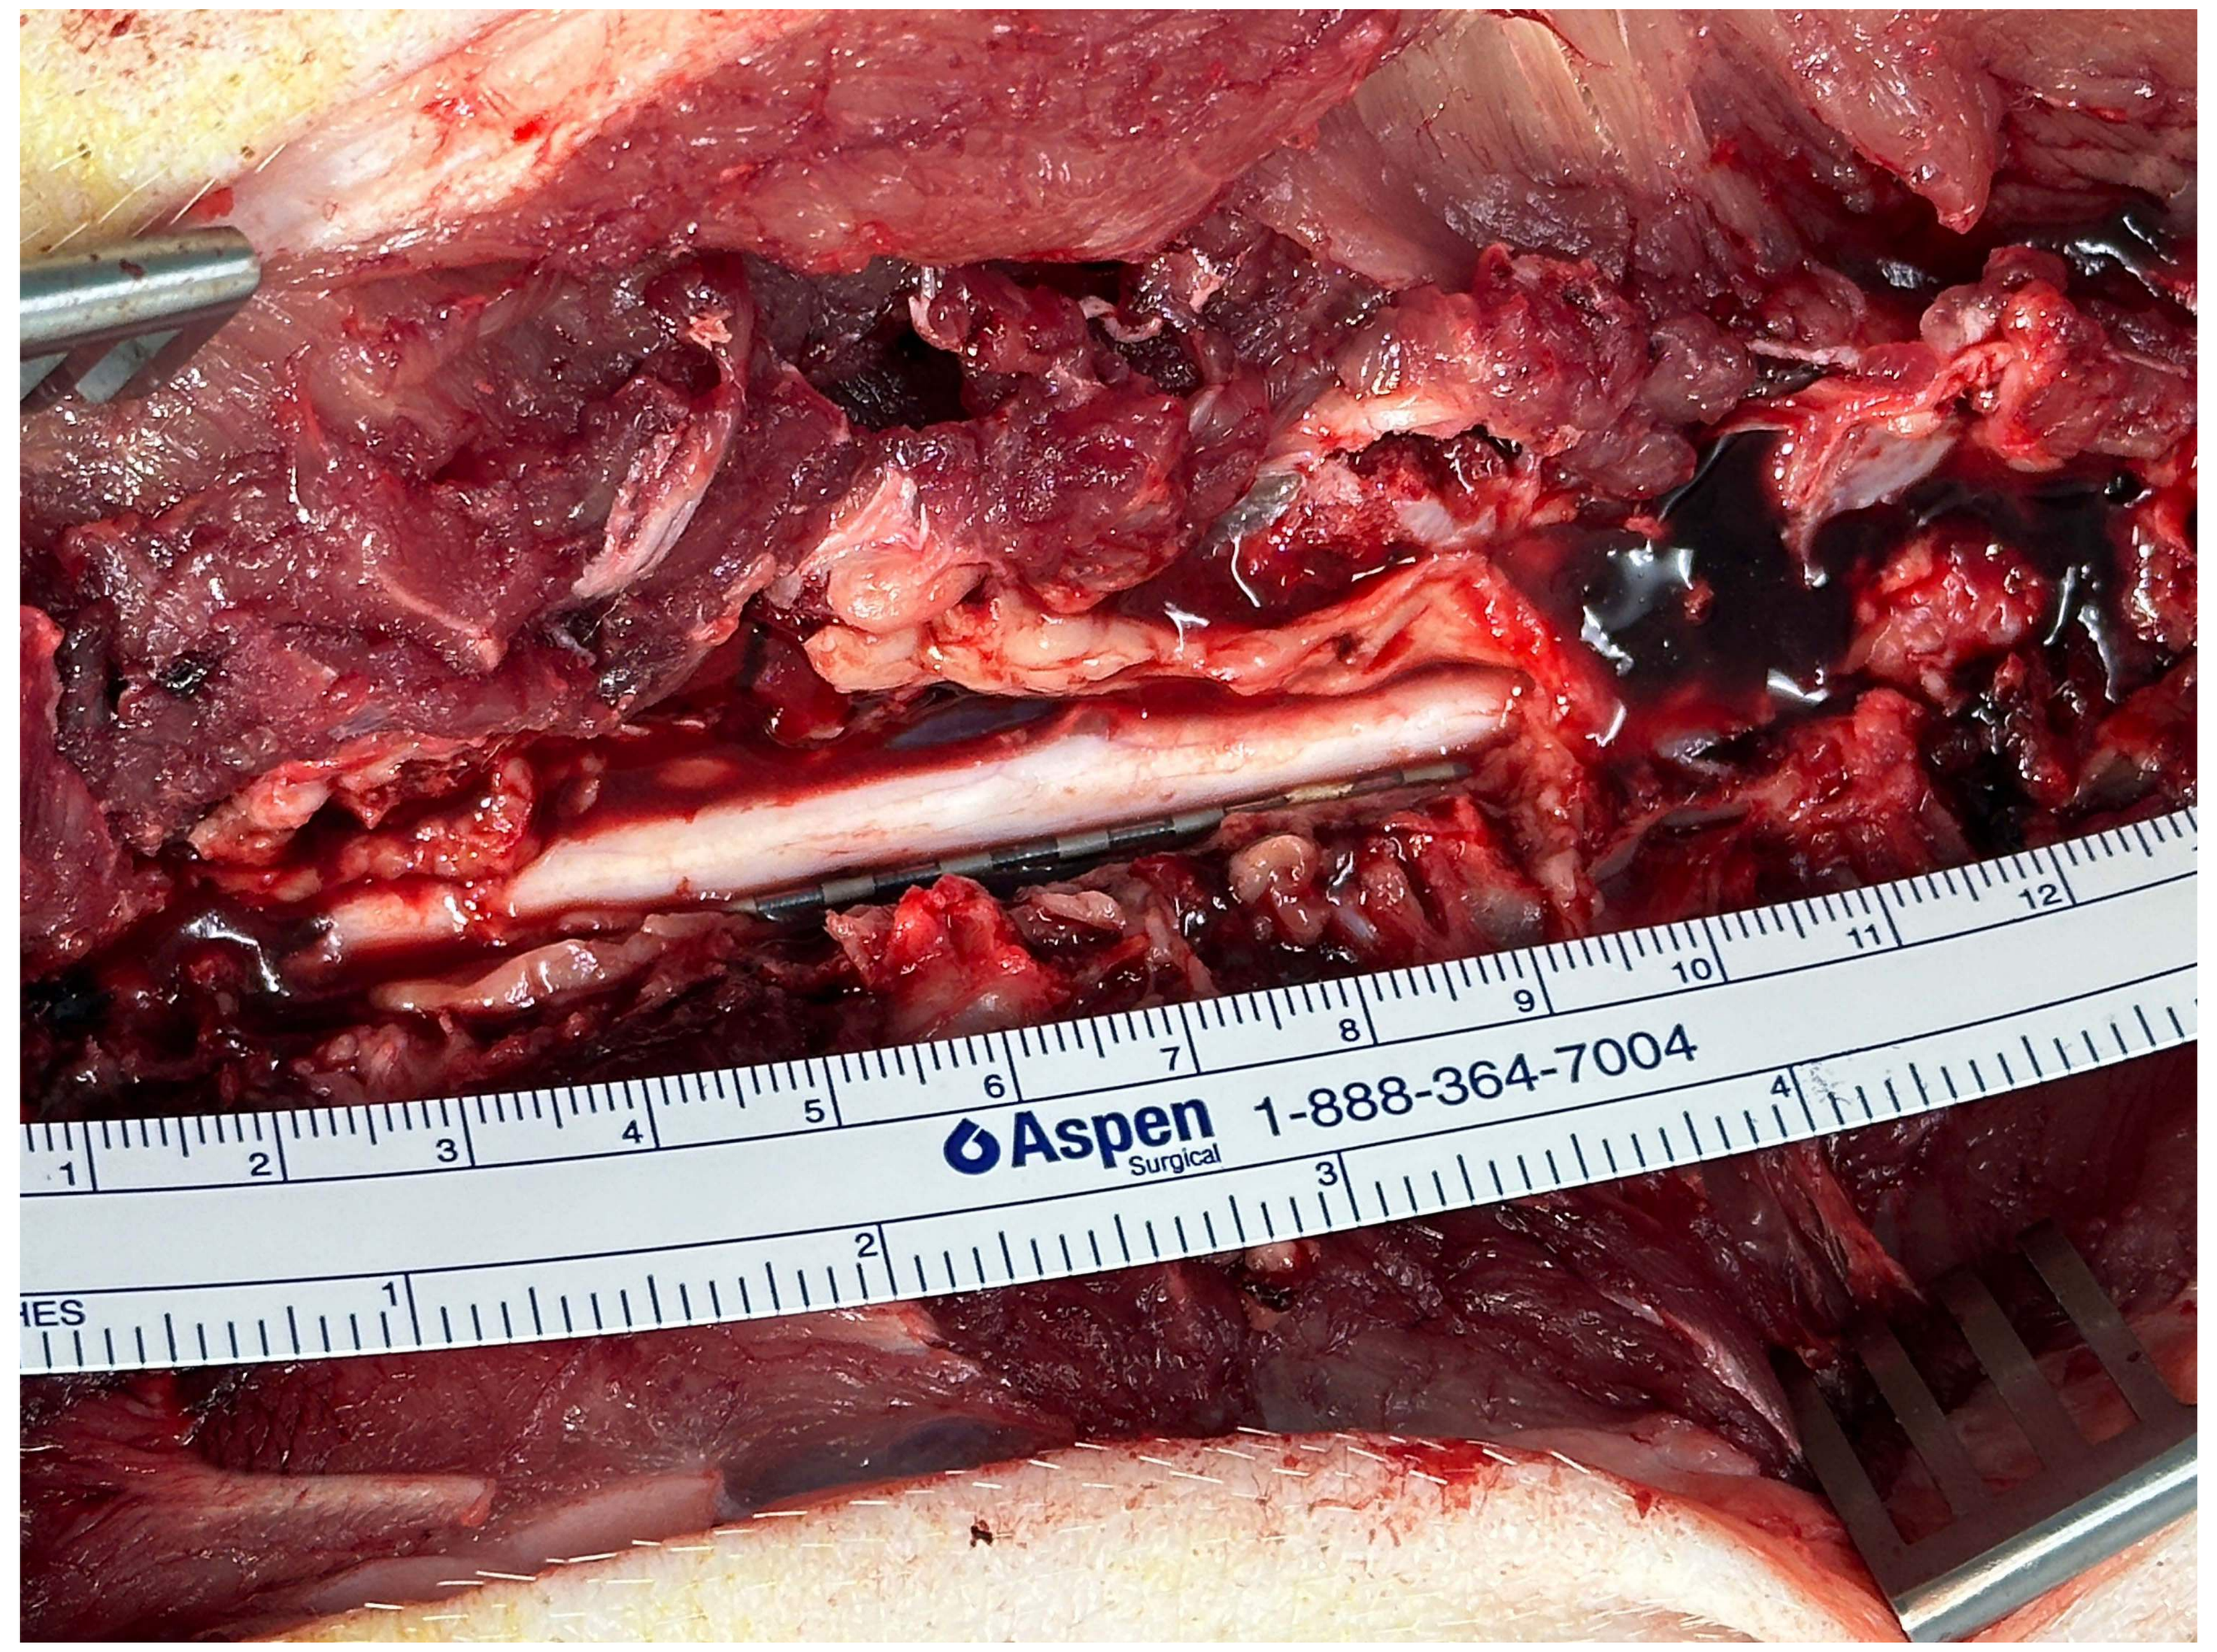
Aspen Surgical
1-888-364-7004

Figure C: Necropsy image of animal 3 demonstrating successful placement of sensor within the epidural space.

Discussion:

- The results from glucose tolerance tests demonstrated that the test device reliably tracked glucose measurements similar to the blood and interstitial fluid
- The device was placed using a percutaneous minimally invasive procedure and time for implant was less than 1 hour
- The animals recovered well with no neurological or physical complications associated with the procedure
- All animals healed well and no device-related adverse events occurred during the 30-60 day time points

Conclusion:

The preliminary findings suggest that glucose sensing in the epidural space is feasible and reliable. This novel approach holds significant promise for simplifying diabetes device management when continuous glucose monitoring is integrated with SCS for people with Painful Diabetic Neuropathy. Further research is needed to assess long-term sensor performance.

Learn more about Glucotrack

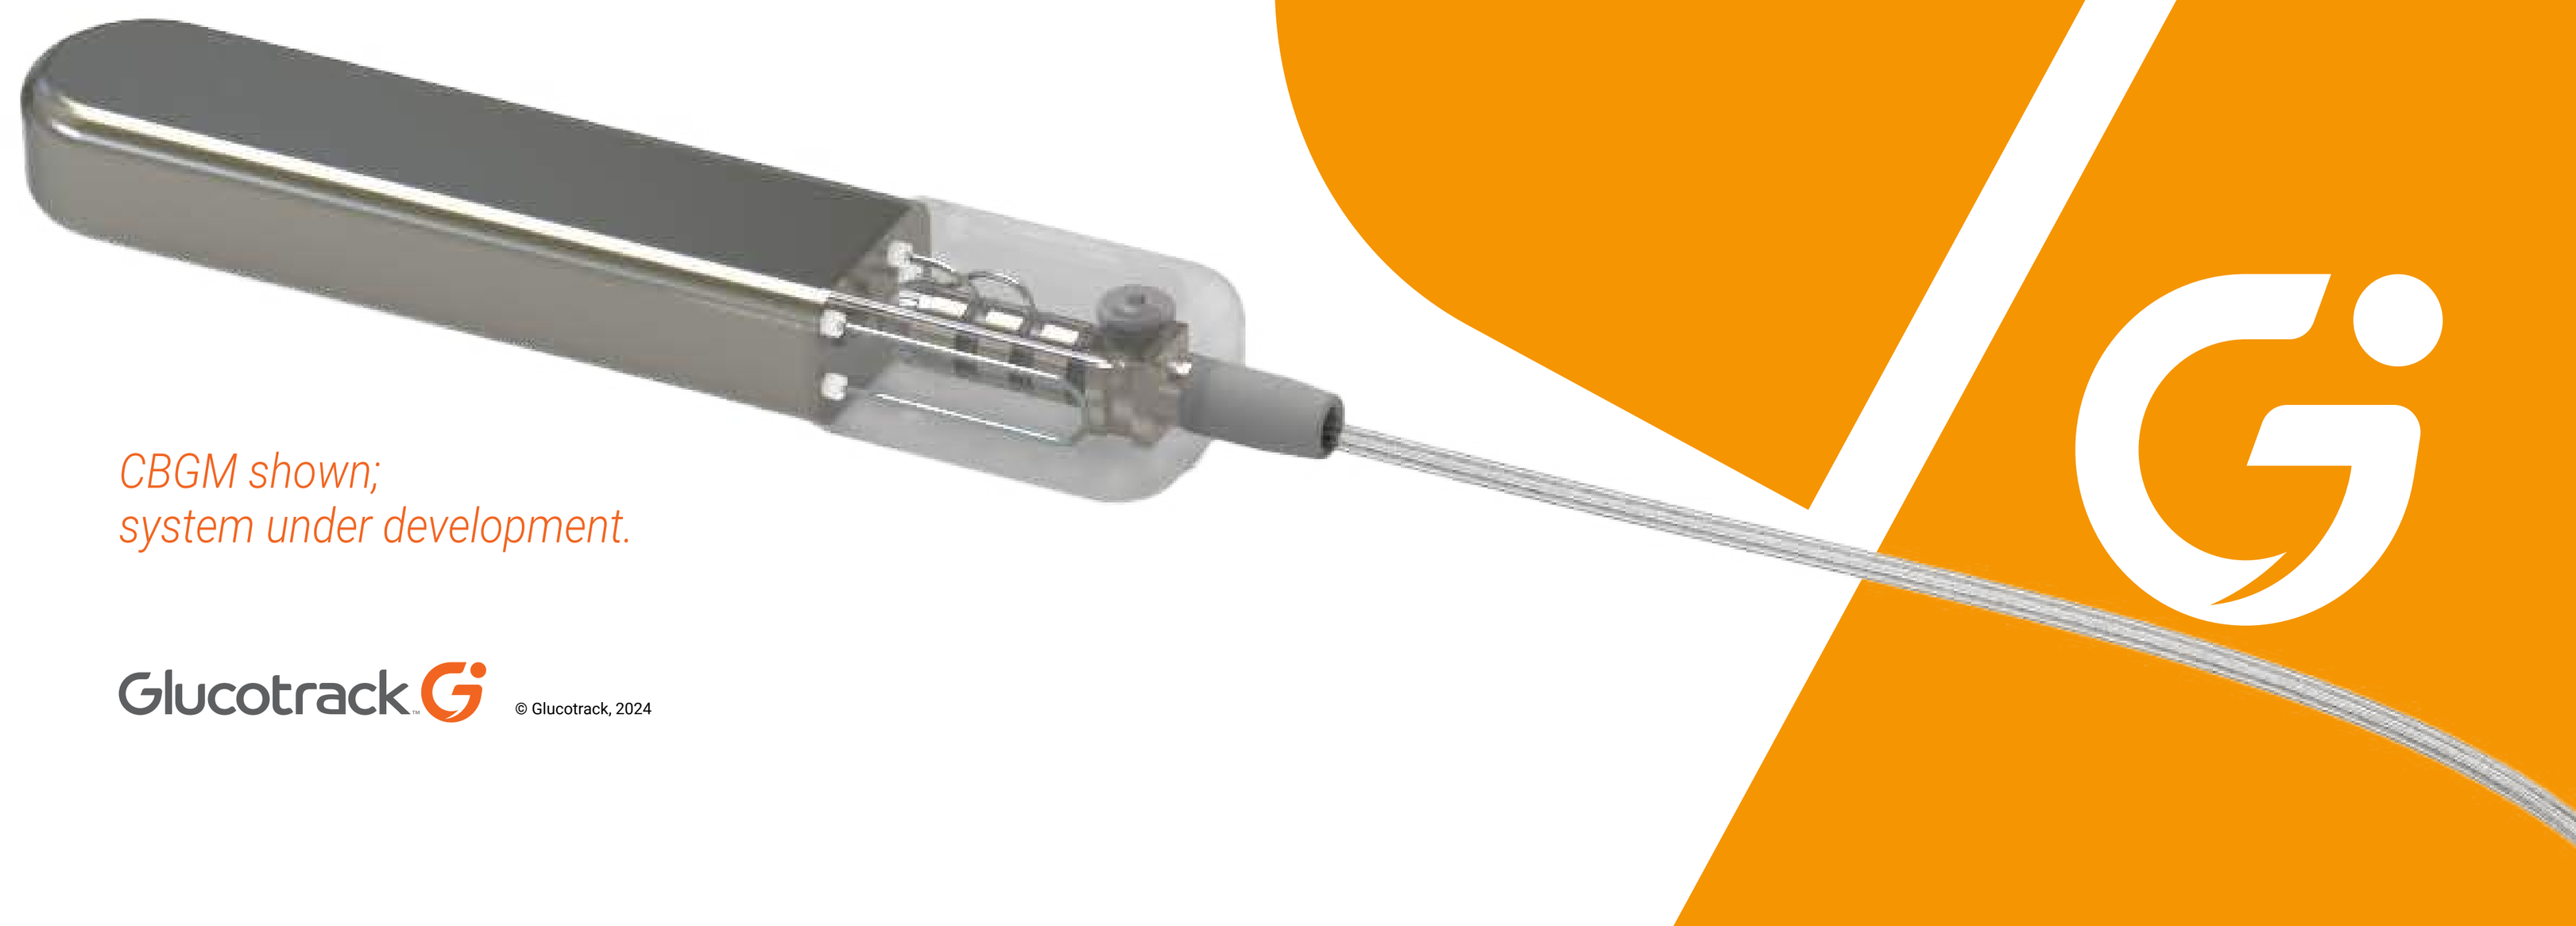

CBGM shown; system under development.

Glucotrack © Glucotrack, 2024