In Vivo Evaluation of Novel Long-Term Intravascular Implantable Continuous Blood Glucose Monitor in a Chronic Ovine Model: A Glucotrack™ Inc. Investigation

Paul V. Goode, PhD; Mark A. Tapsak, PhD; Michael Talcott, D.V.M.; JP Thrower, PhD; Timothy L. Routh, M. Eng.; Stephen Tapsak; Jose Garcia | Corresponding author: Mark A. Tapsak; mtapsak@glucotrack.com

1010-P

Background:

This study provides the first in vivo evaluation of Glucotrack's novel long-term Continuous Blood Glucose Monitor (CBGM) in a chronic ovine model. This investigation aims to understand the practical applications and reliability of the device in a live animal setting to anticipate its performance in humans.

Objective:

The primary objectives of this study were to evaluate the performance of the CBGM, refine placement techniques, and assess chronic gross biocompatibility through explant and necropsy examinations.

Methods:

Two adult male sheep had sensors (2 per sheep) placed intravascularly within the left and right jugular veins. Sheep were chosen as the large animal model due to their large jugular veins, which present flow rates similar to human vessels, facilitating better clinical translation. The glucose metabolism in sheep is well-documented. They are amenable to blood-sampling–related handling, and are a species with well-understood procedures (anesthesia, surgery, and post-operative care), making them ideal for studies involving catheter leads and sensors. Sensor placement was confirmed fluoroscopy [Figure 1A]. The devices measured blood glucose levels at 10-minute intervals regularly and at 2-minute intervals during intravenous glucose tolerance tests (IVGTT) on days 30* and 60* post-sensor placement. Blood samples were taken with a standard Blood Glucose (BG) Monitor during IVGTTs and approximately twice weekly for comparative analysis. Post sensor removal, the raw data was extracted from the memory contained in each device and analyzed.

Data analysis: To evaluate sensor performance, outliers in triplicate BG measurements were eliminated using Dixon's Q Test (critical value 0.970, 95% confidence) and the remaining data averaged. Each average was then matched with the closest preceding sensor signal based on synchronized timestamps.

During the IVGTTs on days 30 and 60, the sensors (2 per animal) were calibrated using linear regression of paired raw signal and BG values to find the optimal slope and intercept. For each reference BG value, the prior calibrated sensor reading was paired for comparison. Mean absolute relative differences (MARDs) were calculated and documented for each IVGTT. Continuous sensor data and BG reference values were plotted for qualitative assessment.

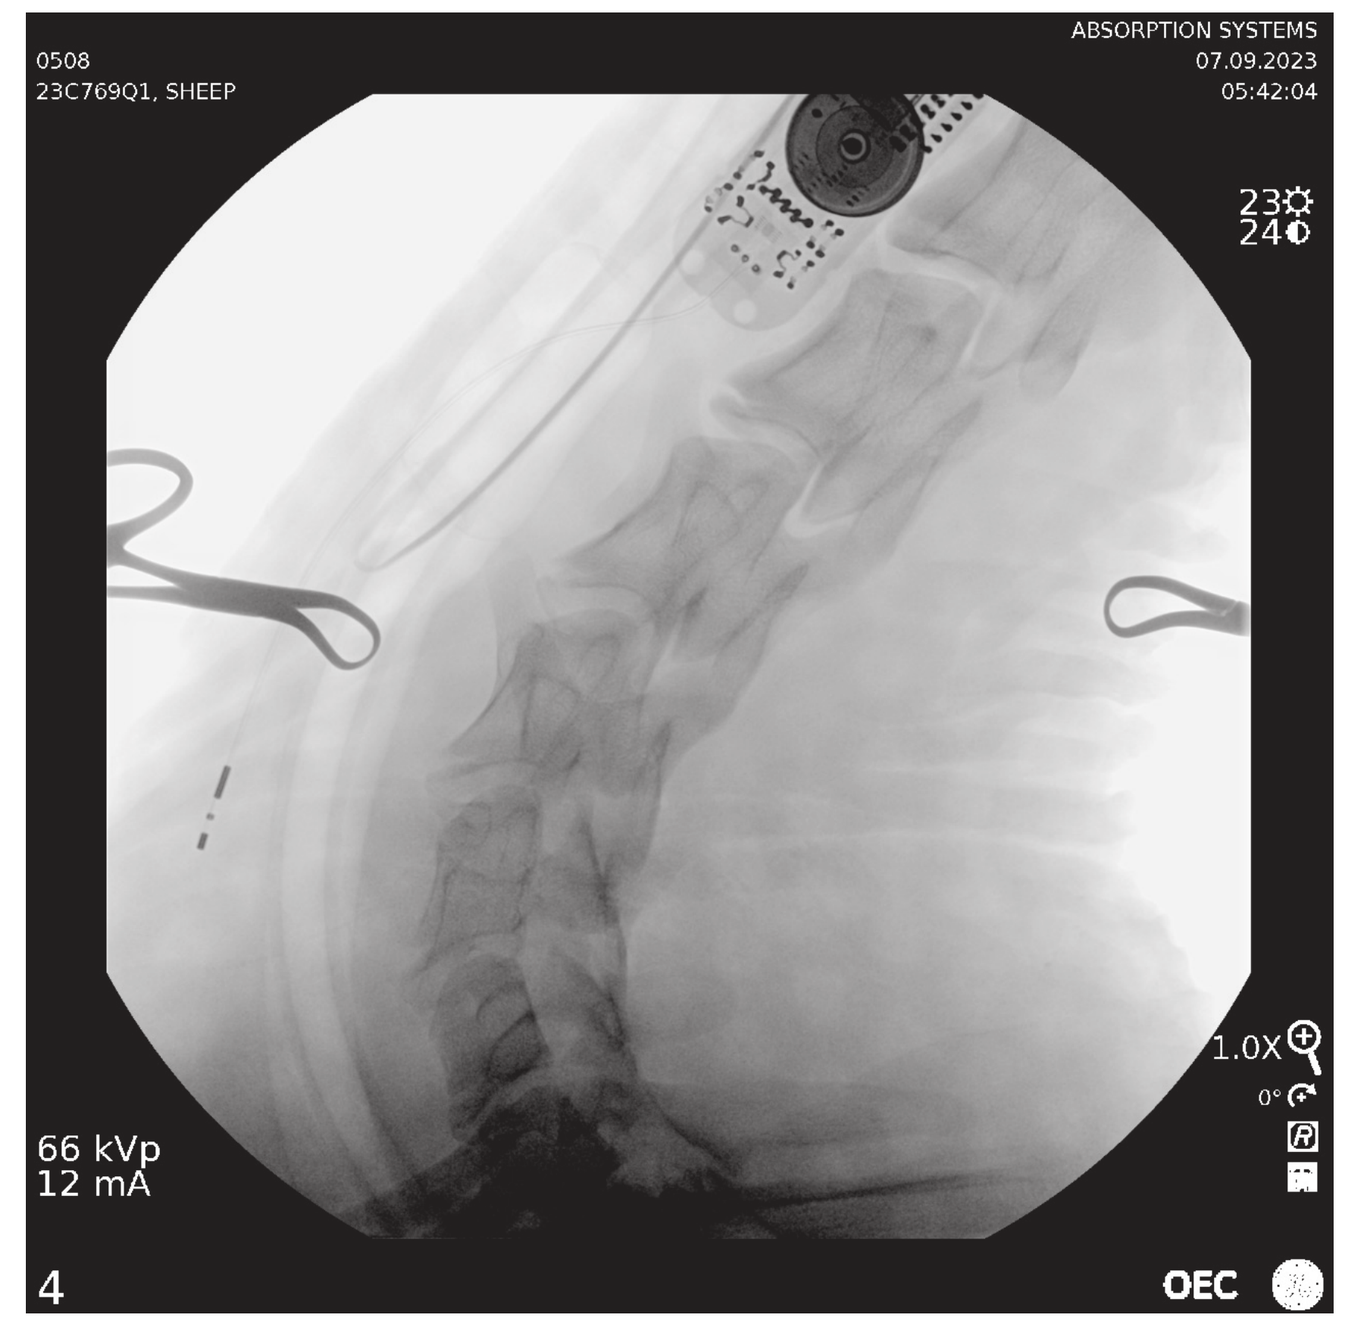
0508
23C769Q1, SHEEP
ABSORPTION SYSTEMS
07.09.2023
05:42:04
66 kVp
12 mA
1.0X
OEC
4

Calibration durability varied among sensors; however, extended calibration durability was demonstrated with 1 sensor. The Day-60 IVGTT calibration for sensor Sheep 1, Right position was maintained for 8 weeks, without adjustments. BG reference values from bi-weekly samples were paired with the latest calibrated sensor readings and MARD was calculated and reported for this period. Plots of 8 continuous weeks of sensor data and reference BG values, including IVGTT results, are shown to the right.

*Actual Day 32 and 57.

[Figure 1A]: Fluoroscopy image of the sheep's neck region. The device can be seen at the top, sensor electrodes are toward the bottom left.

Results:

- Successful placement was achieved within approximately 20 minutes without the need for customized tools
- The MARD decreased from an average of 9.3% on Day 30 to 4.3% on Day 60, indicating improved accuracy over time for each sensor
- Performance plots illustrate the sensor accuracy and reliability over the study period
- No serious adverse events were observed throughout the study

MARD Table - All Intravenous Sensors - Calibrated With IVGTT BG

Sensor ID:	Day 30 IVGTT MARD	Day 60 IVGTT MARD
Sheep 1, Left	7.1%	6.0%
Sheep 1, Right	7.9%	3.4%
Sheep 2, Left	10.6%	5.8%
Sheep 2, Right	11.5%	1.8%
Average:	**9.3%**	**4.3%**

Day-30 IVGTT Plots:

Day-30 IVGTT - Sheep #1

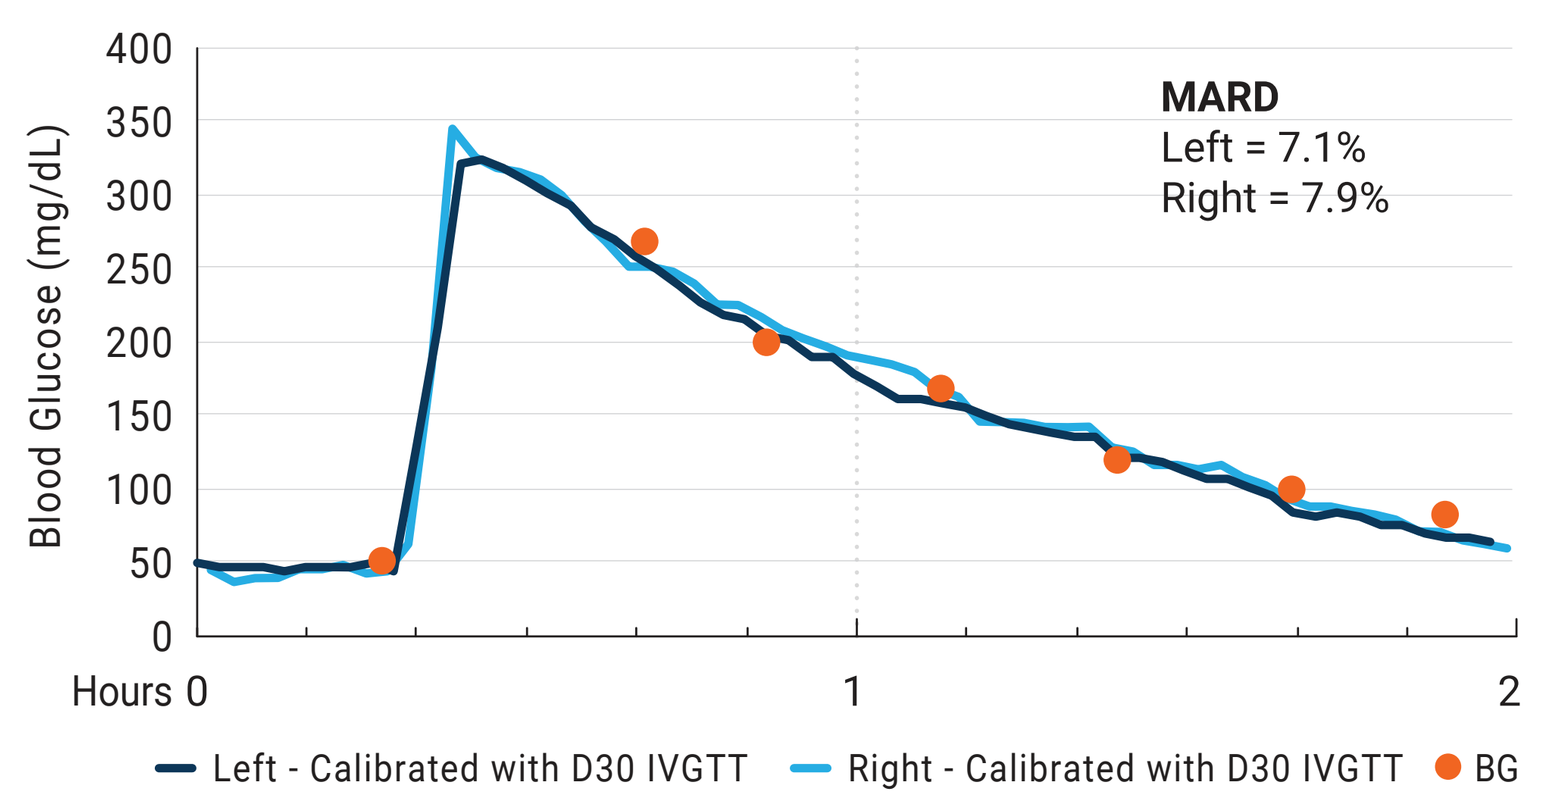
Blood Glucose (mg/dL)
Hours
MARD
Left = 7.1%
Right = 7.9%
Left - Calibrated with D30 IVGTT
Right - Calibrated with D30 IVGTT
BG

Day-30 IVGTT - Sheep #2

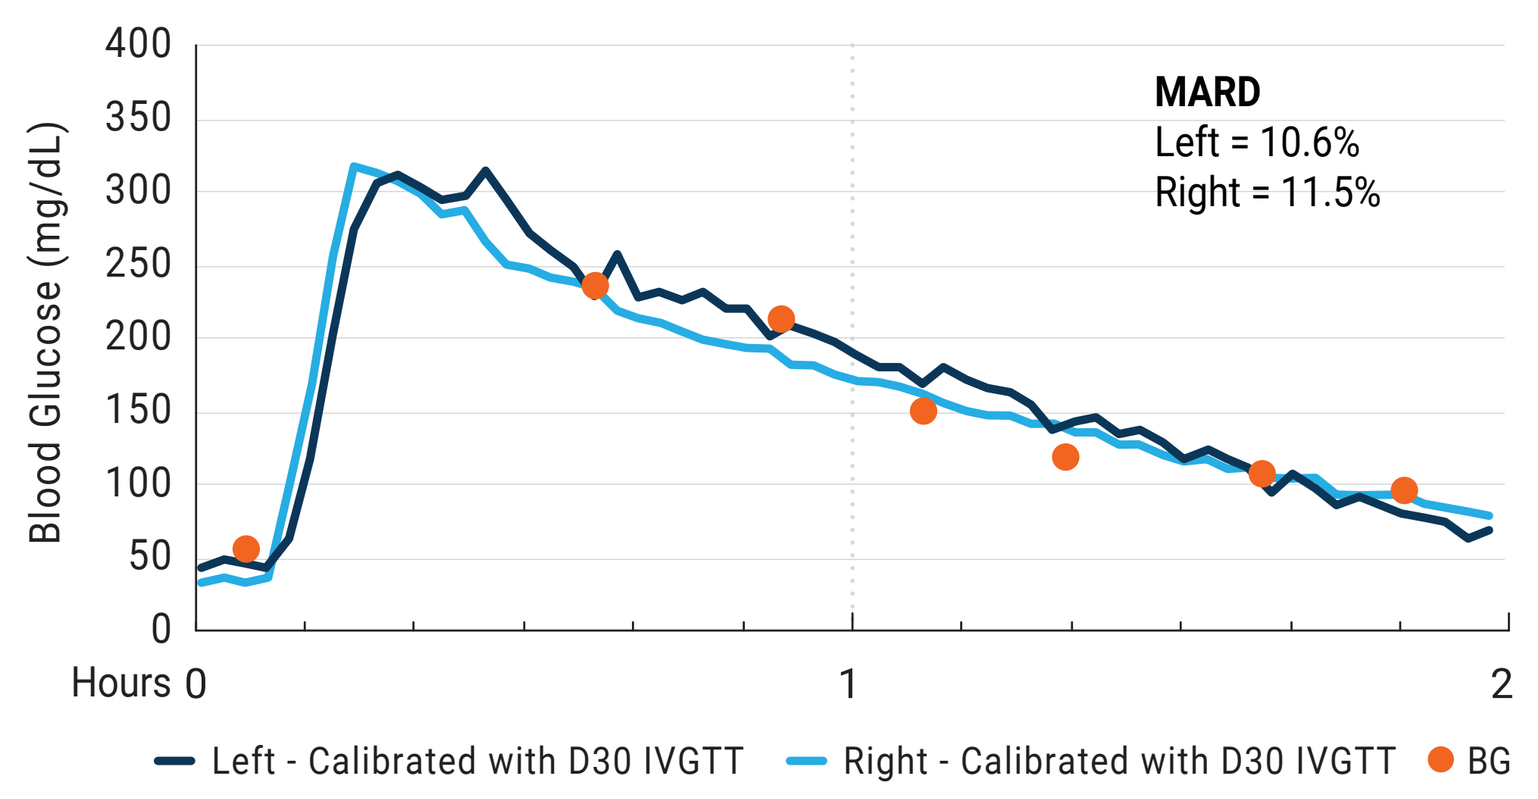
Blood Glucose (mg/dL)
Hours
MARD
Left = 10.6%
Right = 11.5%
Left - Calibrated with D30 IVGTT
Right - Calibrated with D30 IVGTT
BG

Day-60 IVGTT Plots

Day-60 IVGTT - Sheep #1

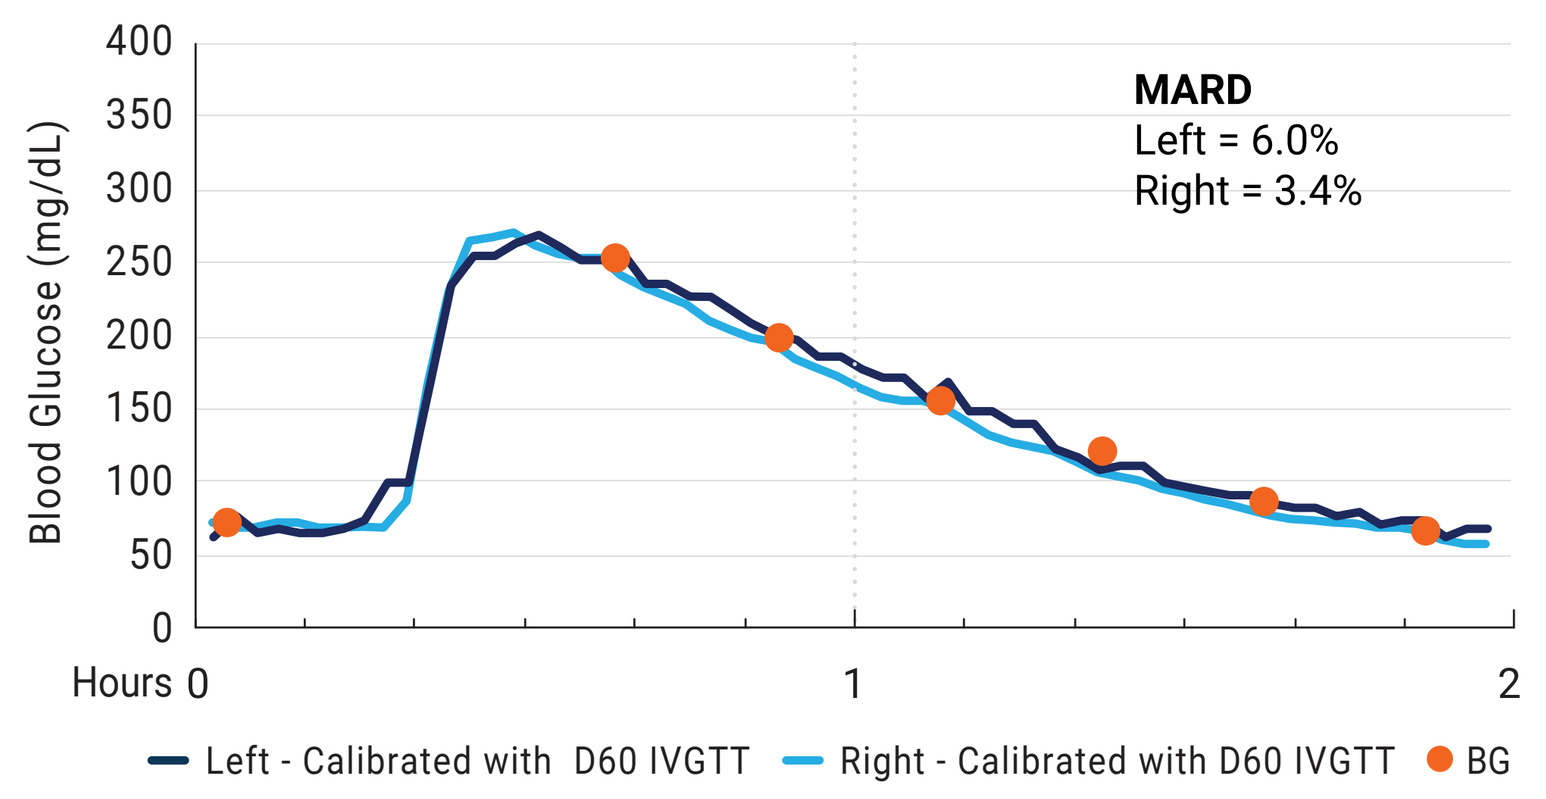
Blood Glucose (mg/dL)
Hours
MARD
Left = 6.0%
Right = 3.4%
Left - Calibrated with D60 IVGTT
Right - Calibrated with D60 IVGTT
BG

Day-60 IVGTT - Sheep #2

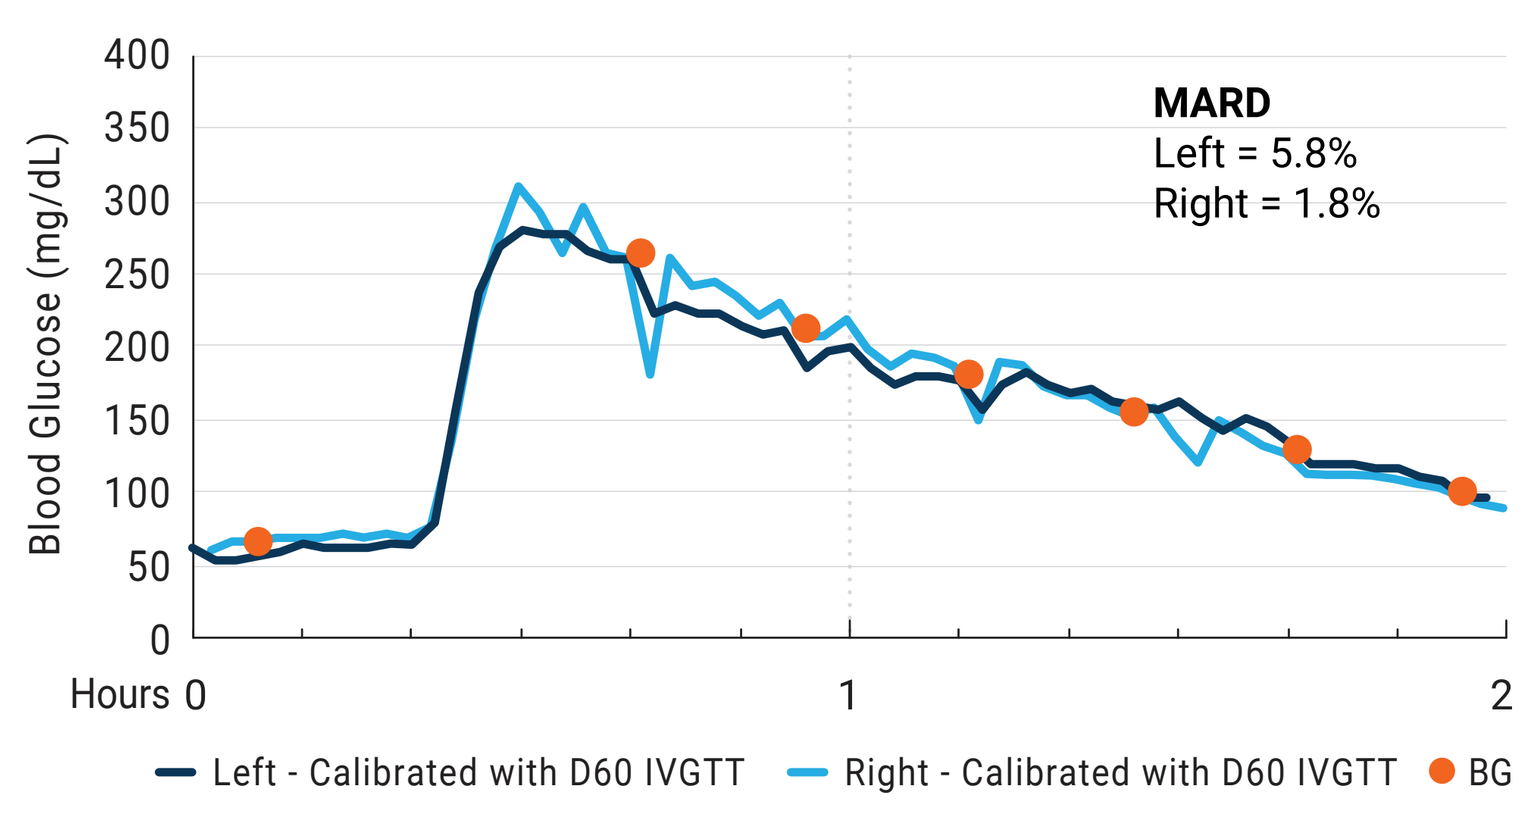
Blood Glucose (mg/dL)
Hours
MARD
Left = 5.8%
Right = 1.8%
Left - Calibrated with D60 IVGTT
Right - Calibrated with D60 IVGTT
BG

8-Week Calibration Durability Example:

11.6% MARD - sensor Sheep 1, Right, over 8-week period, calibrated with Day-60 (actual Day-57) IVGTT, with interim twice-weekly BG as reference (no recalibration), 4 weeks before calibration and 4 weeks after

Sheep #1 - Sensor Sheep 1, Right - 8 Weeks With Single Calibration - (4 Weeks Before Calibration - Day 29 through 84 - Calibration on Day 57)

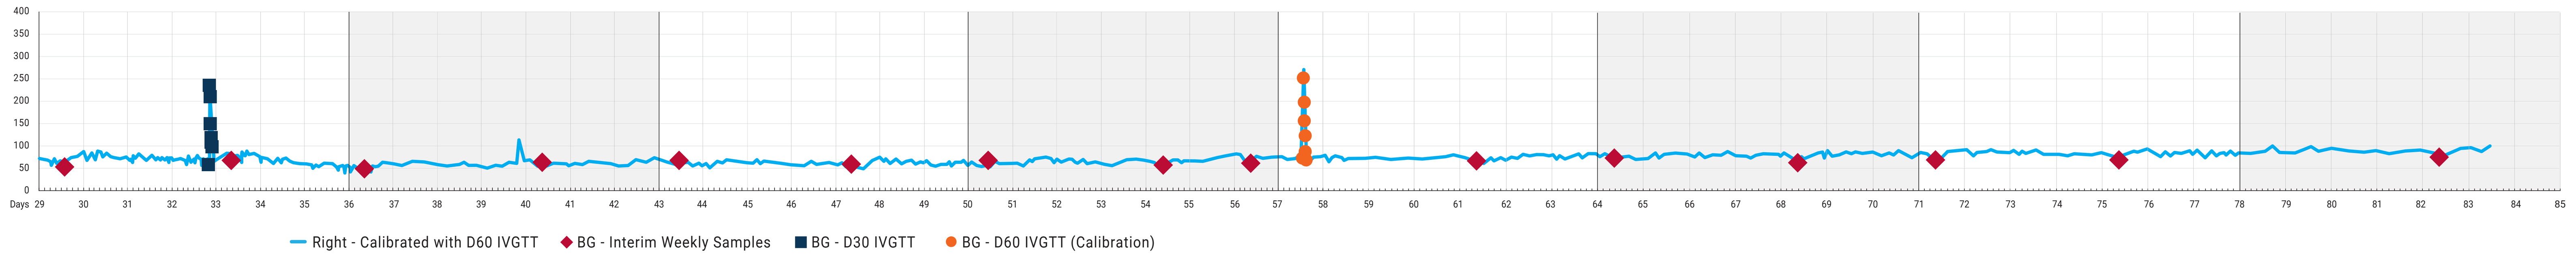
Days
Right - Calibrated with D60 IVGTT
BG - Interim Weekly Samples
BG - D30 IVGTT
BG - D60 IVGTT (Calibration)

Discussion:

- Sensors consistently tracked blood glucose accurately and remained linear during Day-30 and Day-60 IVGTTs
- The raw glucose signal was smooth with a high signal-to-noise ratio and minimal outliers, supporting potential algorithm improvements
- The sensors rapidly and accurately responded to significant glucose infusions, demonstrating the advantages of direct-in-blood measurements
- Multi-week calibration stability indicates that CBGM technology can operate for extended periods with minimal recalibration
- Sheep, with a blood glucose range of 50-80 mg/dL, effectively allowed for sensor evaluation within CGMs' typical sensing range of 40-400 mg/dL, especially at lower levels around 60 mg/dL
- MARD from Day-30 and Day-60 IVGTTs indicates that sensor performance remained stable throughout the study
- Pathology results post sensor removal were unremarkable, confirming the safety and biocompatibility of the device

Conclusion:

The initial animal trials of the Glucotrack CBGM device demonstrated a high correlation with glucose levels, significant long-term stability, and consistent performance, suggesting its potential as a reliable and accurate long-term glucose monitoring solution for humans. The ovine model proved suitable for evaluating in-vessel CBGM devices, providing a stable environment for detailed performance assessment.

Learn more about Glucotrack

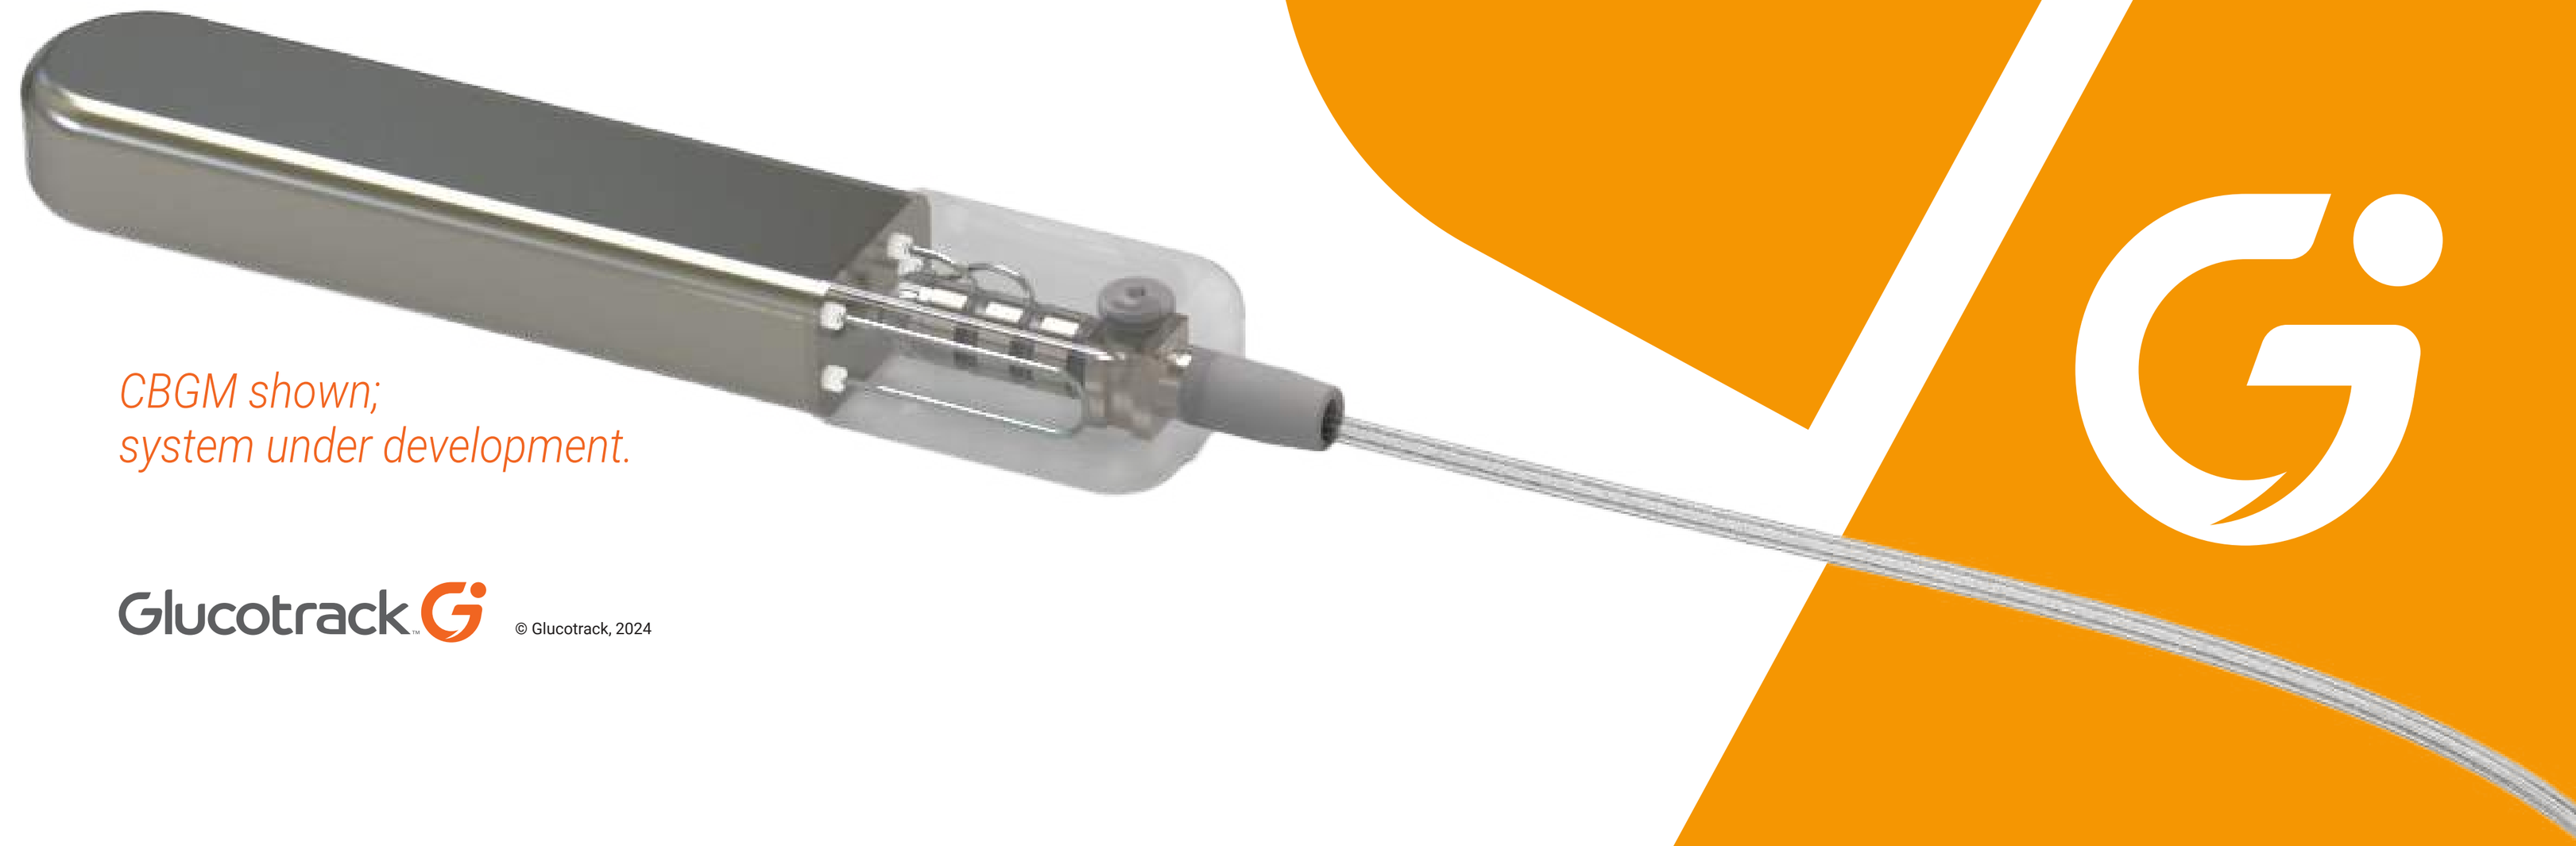

CBGM shown; system under development.

Glucotrack © Glucotrack, 2024